Macaque rhadinovirus-associated B-cell lymphomas in cynomolgus monkeys following long-term exposure to an immunomodulatory biotherapeutic

Sébastien Laurent[1], Ambroise Garry[2], Frédéric Gervais[3], Svenja R. Hartung[4], Cris Kamperschroer[5], Marc Niehoff[4], Annette Romeike[4], Carolin Schramm[6], Fatima Tariq[2]

[1]In vivo Operations, Preclinical Safety, Sanofi, Montpellier, France, [2]Investigative Toxicology, Preclinical Safety, Sanofi, Vitry sur Seine, France, [3]Charles River Laboratories Evreux, Miserey, France, [4]Labcorp, Münster, Germany, [5]Investigative Toxicology, Preclinical Safety, Sanofi, Cambridge, USA, [6]R&D Projects EU, Sanofi, Cambridge, Great Britain

BACKGROUND & INTRODUCTION

- Repeat dosing of non-human primates (NHPs) with immunomodulatory drugs in the context of drug safety evaluation may be associated with opportunistic infections.
- **Rhadinoviruses** (RV) are gamma-2 herpesviruses; they include the Kaposi's Sarcoma herpesvirus (RV-1 lineage) in humans and some important viruses of NHPs, *i.e.*, retroperitoneal fibromatosis-associated herpesvirus (RV-1 lineage) and rhesus rhadinovirus (RRV; RV-2 lineage).
- Infection with RRV commonly occurs early in life but is typically asymptomatic in immunocompetent animals.
- In contrast, SIV-coinfected animals develop marked B cell lymphocytosis, and experimental inoculation with RRV has been associated with a lymphoproliferative disorder resembling multicentric Castleman's disease.

Figure 1. Phylogeny of gammaherpesviruses

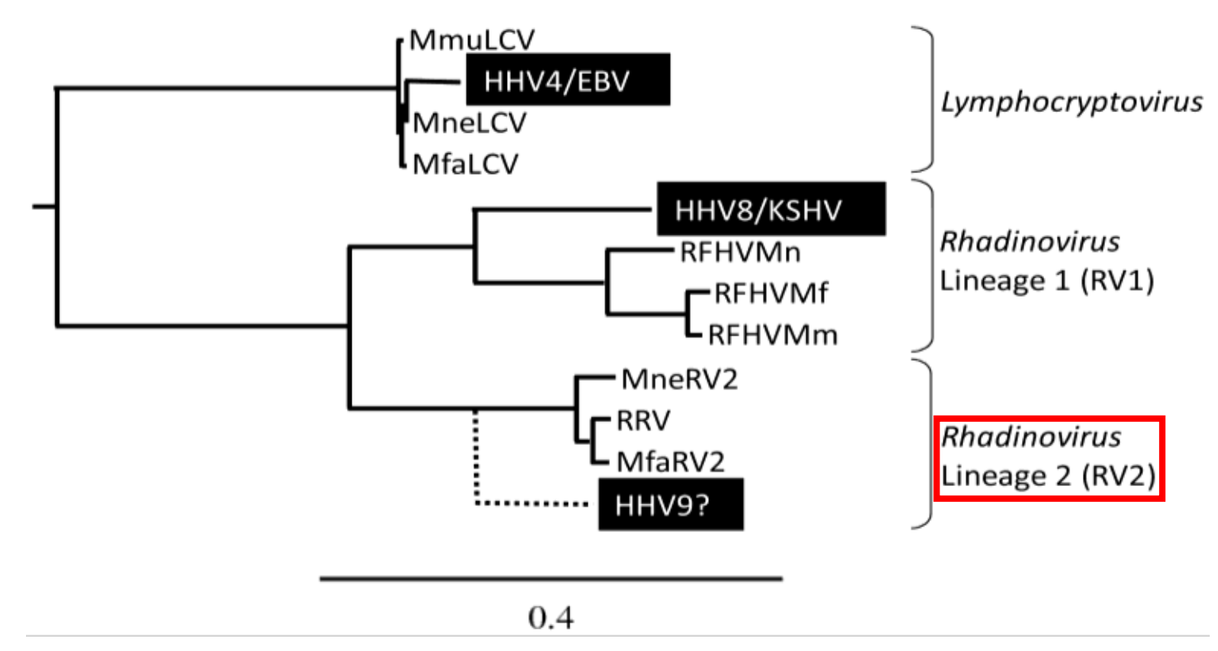
MmuLCV
HHV4/EBV
MneLCV
MfaLCV
Lymphocryptovirus
HHV8/KSHV
RFHVMn
RFHVMf
RFHVMm
Rhadinovirus Lineage 1 (RV1)
MneRV2
RRV
MfaRV2
HHV9?
Rhadinovirus Lineage 2 (RV2)
0.4

Table 1: Gammaherpesviruses of interest in macaques

Macaque virus	Closely related virus in humans	Genus / lineage
Macaque LCV	EBV (HHV4)	Lymphocryptovirus
Macaque RFHV	KSHV (HHV8)	Rhadinovirus / RV-1 lineage
RRV	HHV9?	Rhadinovirus / RV-2 lineage

EBV: Epstein-Barr Virus; HHV: Human herpesvirus; KSHV: Kaposi's sarcoma herpesvirus; LCV: Lymphocryptovirus; Mfa: *M. fascicularis*; Mm: *M. mulatta*; Mne: *M. nemestrina;* RFHV: Retroperitoneal fibromatosis herpesvirus; RV: Rhadinovirus; RRV: Rhesus Rhadinovirus.

- A **6-month subcutaneous repeat-dose toxicity study** was conducted in **cynomolgus monkeys** (*Macaca fascicularis*) with an **immunomodulatory biotherapeutic.** Hematolymphoid tumors were observed in some high dose animals, leading to several investigations aimed at addressing their etiology and pathogenesis.

MATERIAL & METHODS

- **Chronic subcutaneous toxicity study in the NHP**
 - Immunomodulatory biotherapeutic given once weekly at two dose levels for 6 months; 12-week recovery period
 - Cynomolgus monkeys (Mauritius), sexually mature
 - Clinical pathology & immunophenotyping on peripheral blood at predose, Week 13 and Week 26

Table 2: Group assignment

Group	Number of animals	
	Males	Females
1 (Control)*	6	6
2 (Low Dose)	4	4
3 (High dose)*	6	6

*2 animals/sex/group in Groups 1 and 3 designated as recovery animals

- **Immunohistochemistry (IHC) for LCV EBNA2**
 - FFPE blocks; Ventana Discovery Ultra 1
 - Mouse anti-EBNA2 monoclonal antibody (ab90543, Abcam); CC1 antigen retrieval
 - Positive control: sections of RAJI cells
- **IHC for CD20, CD3, iba-1**
 - FFPE blocks; manual assays
 - Rabbit anti-iba-1 polyclonal antibody (PA5-2127, Invitrogen), mouse monoclonal anti-CD20 antibody, rabbit anti-CD3 polyclonal antibody (M0755 & A0452, Dako); heat antigen retrieval
- ***In situ* hybridization (ISH) for RV and LCV**
 - FFPE blocks; RNAscope®; Ventana Discovery Ultra 1
 - Specific probes: V-cyLCV1-Pol RNAscope 2.5 VS probe and V-RRV RNAscope 2.5 VS probe (ACD)
 - PPIB & dapB as positive and negative controls
- **qPCR for viral loads**
 - Blood samples collected twice in predose and once monthly during treatment
 - DNA isolated and assays run to measure macaque LCV, CMV and RV (cyno RV-2) loads
- **Serology**
 - IgG testing on serum samples at predose for several primate viruses, including RV and LCV

RESULTS

1. *In vivo* phase

- One male and one female given the high dose euthanized prematurely on Day 171 and Day 175
- Body weight reduction, hypoactivity, hunched posture and pale mucosae noted in both animals

2. Necropsy findings

- Enlarged spleen and mesenteric lymph node, and beige, firm nodules in non-lymphoid organs (*e.g.*, liver, lung, heart) in both prematurely sacrificed animals and in one terminally sacrificed male at the high dose

3. Clinical pathology

Table 3: Selected changes in clinical pathology parameters prior to necropsy in both animals euthanized prematurely

Parameter(s)	Male	Female
RBCs, hemoglobin, hematocrit	↓↓ to ↓↓↓	↓↓ or ↓↓↓
C-reactive protein	↑↑	↑↑↑
Fibrinogen	↑↑	↑
Albumin and total protein	↓ to ↓↓	↓↓↓
ALT and AST	NC	↑ to ↑↑
GLDH	↑	↑↑
BUN, creatinine and phosphorus	NC	↑ to ↑↑↑

RBCs: red blood cells; NC: no change

4. Histopathology

- Infiltration of various tissues from those three high dose animals by round to polygonal, monomorphic cells arranged in closely packed sheets, and embedded into minor amounts of pre-existing stroma
- Neoplastic cells: 15 to 25 µm in diameter, with distinct cell borders and moderate amounts of amphophilic cytoplasm and one central, large nucleus with one or two, distinct and prominent nucleoli, displaying significant anisocytosis/anisokaryosis, starry sky pattern, high mitotic ratio, and atypical mitoses

Figure 2. Microscopic aspect of the hematolymphoid tumors

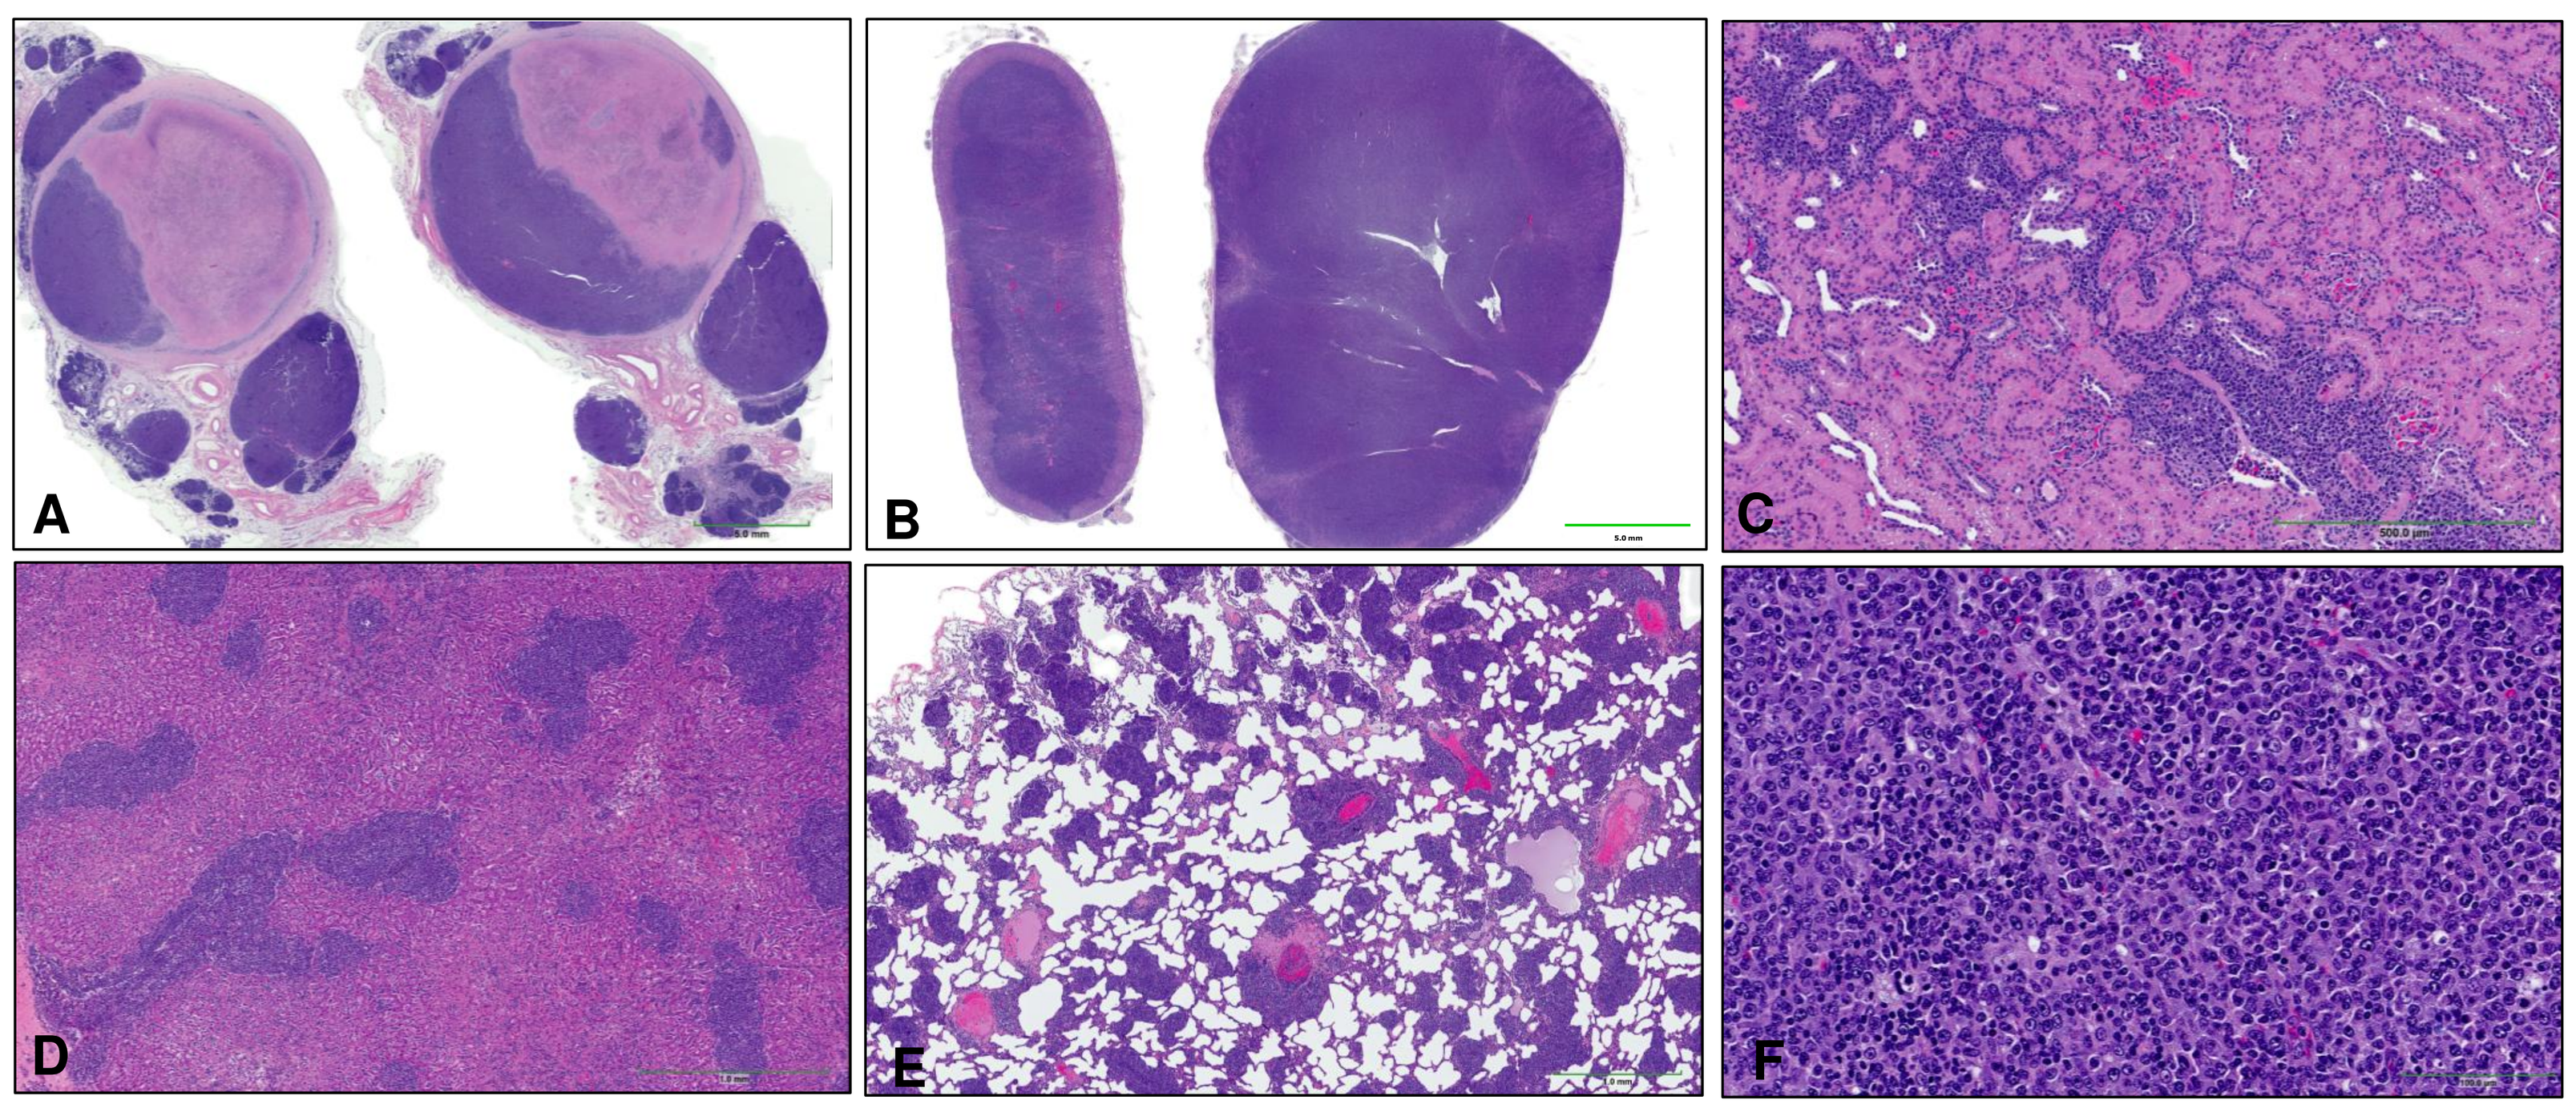
A
B
C
D
E
F

Photomicrographs of various tissues from the high dose female euthanized prematurely. The mesenteric lymph node (A), adrenal glands (B), kidney (C), liver (D) and lung (E) are infiltrated by sheets of neoplastic round cells that disturb their architecture. At high magnification (F), neoplastic cells show prominent cytonuclear atypia and a high mitotic rate. H&E staining.

5. Immunohistochemical characterization of the tumors

- Strong positive membranous staining for CD20 (B-cell marker) of most neoplastic cells
- Negative staining of neoplastic cells for CD3 (T-cell marker) and iba-1 (macrophage marker)
- Low number of infiltrating CD3 positive and iba-1 positive cells
- Clinicopathologic, histopathologic and immunohistochemical profiles consistent with a diagnosis of late-stage **B-cell lymphomas** in three high dose animals

Figure 3. IHC of the hematolymphoid tumors

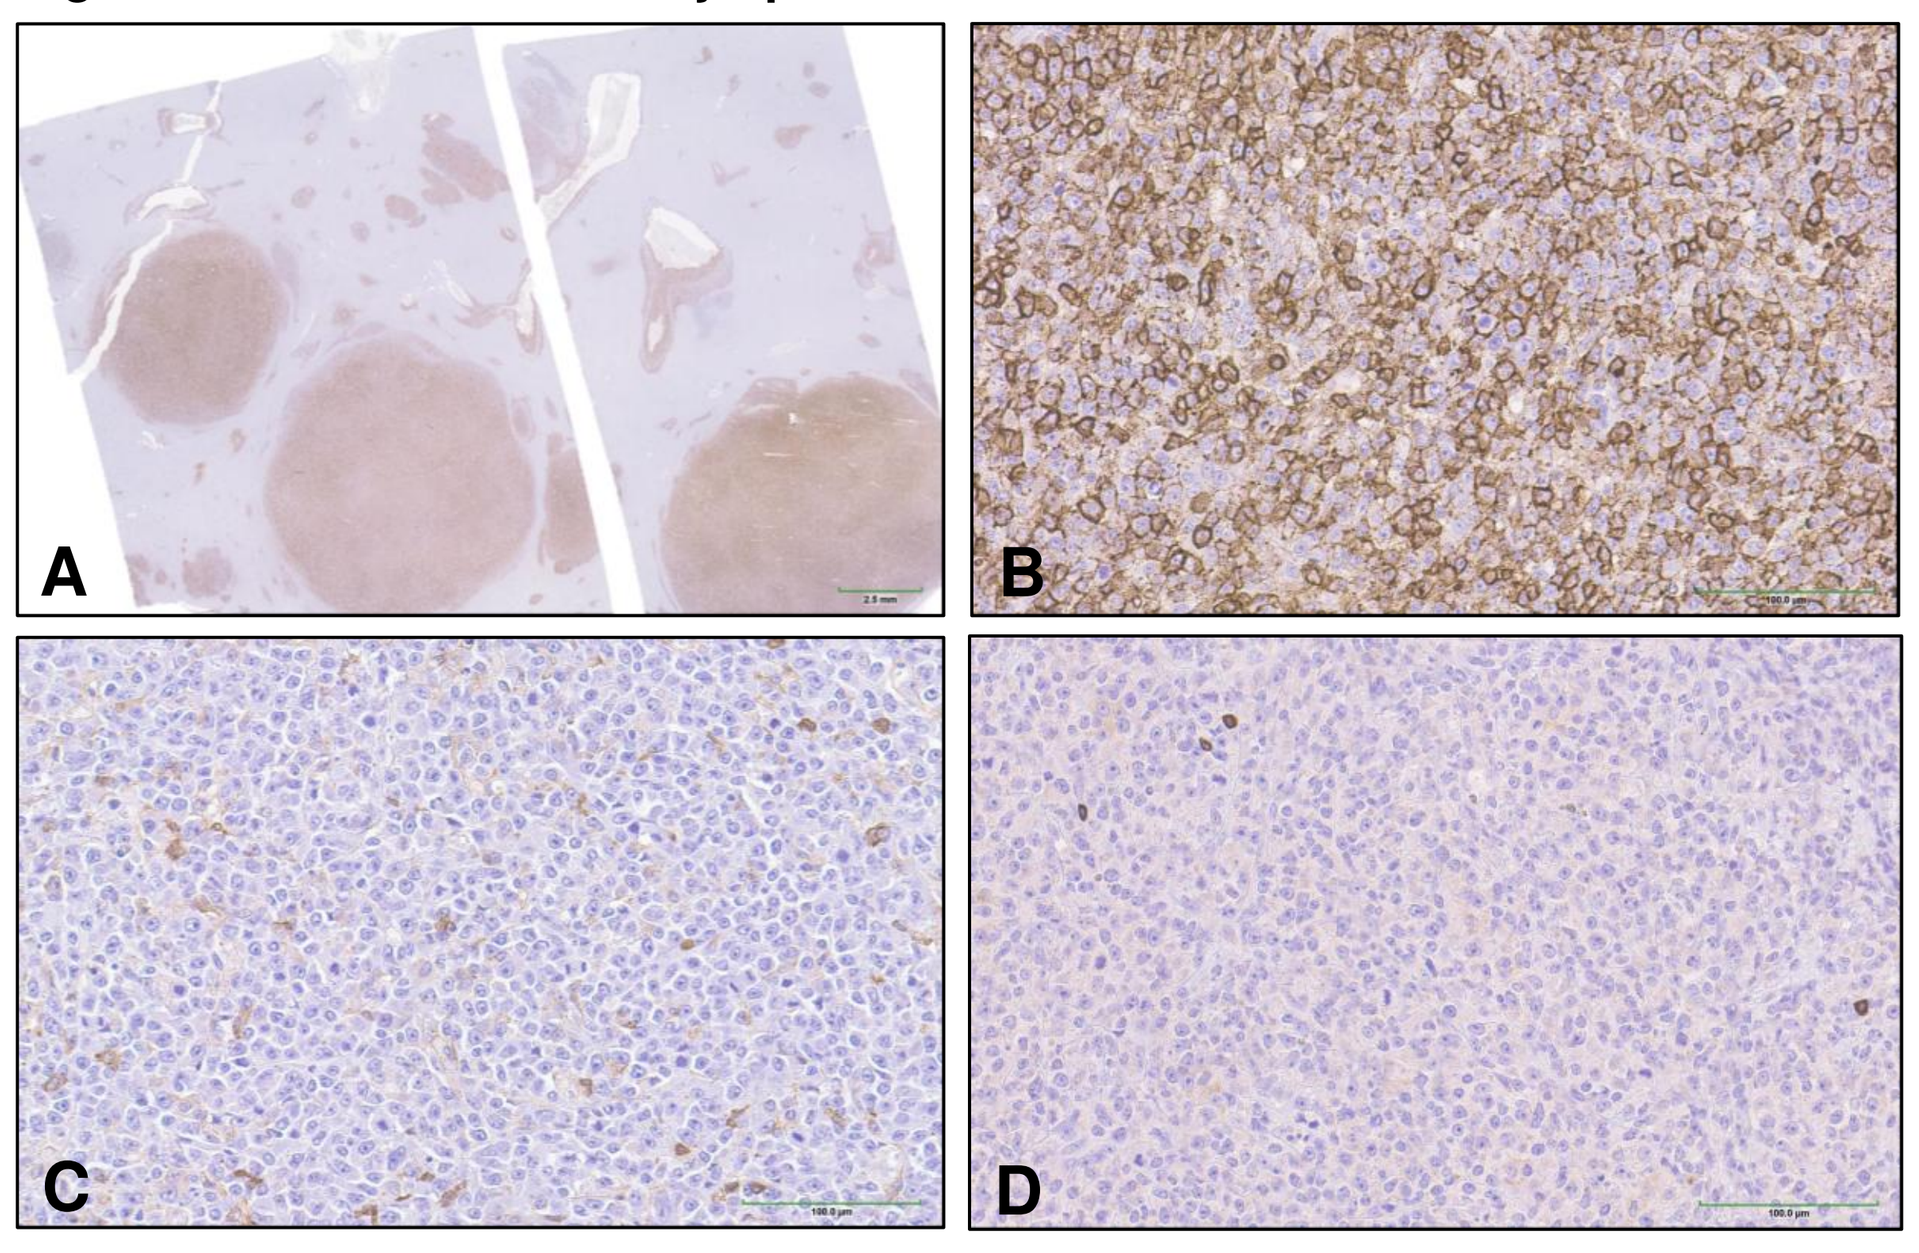
A
B
C
D

IHC for CD20 (A, B), CD3 (C) and iba-1 (D) on the liver, using DAB as chromogen. The tumor is composed of CD20 positive cells, along with rare infiltrating CD3 positive cells and iba-1 positive cells.

RESULTS (CONTINUED)

6. ISH and IHC for gammaherpesviruses in the tumors

- **Strong positive specific staining for RV mRNA in neoplastic cells by ISH**
- No specific staining in neoplastic cells for LCV mRNA by ISH
- Neoplastic cells negative for EBNA2 (EBV nuclear antigen 2) by IHC (*not shown*)

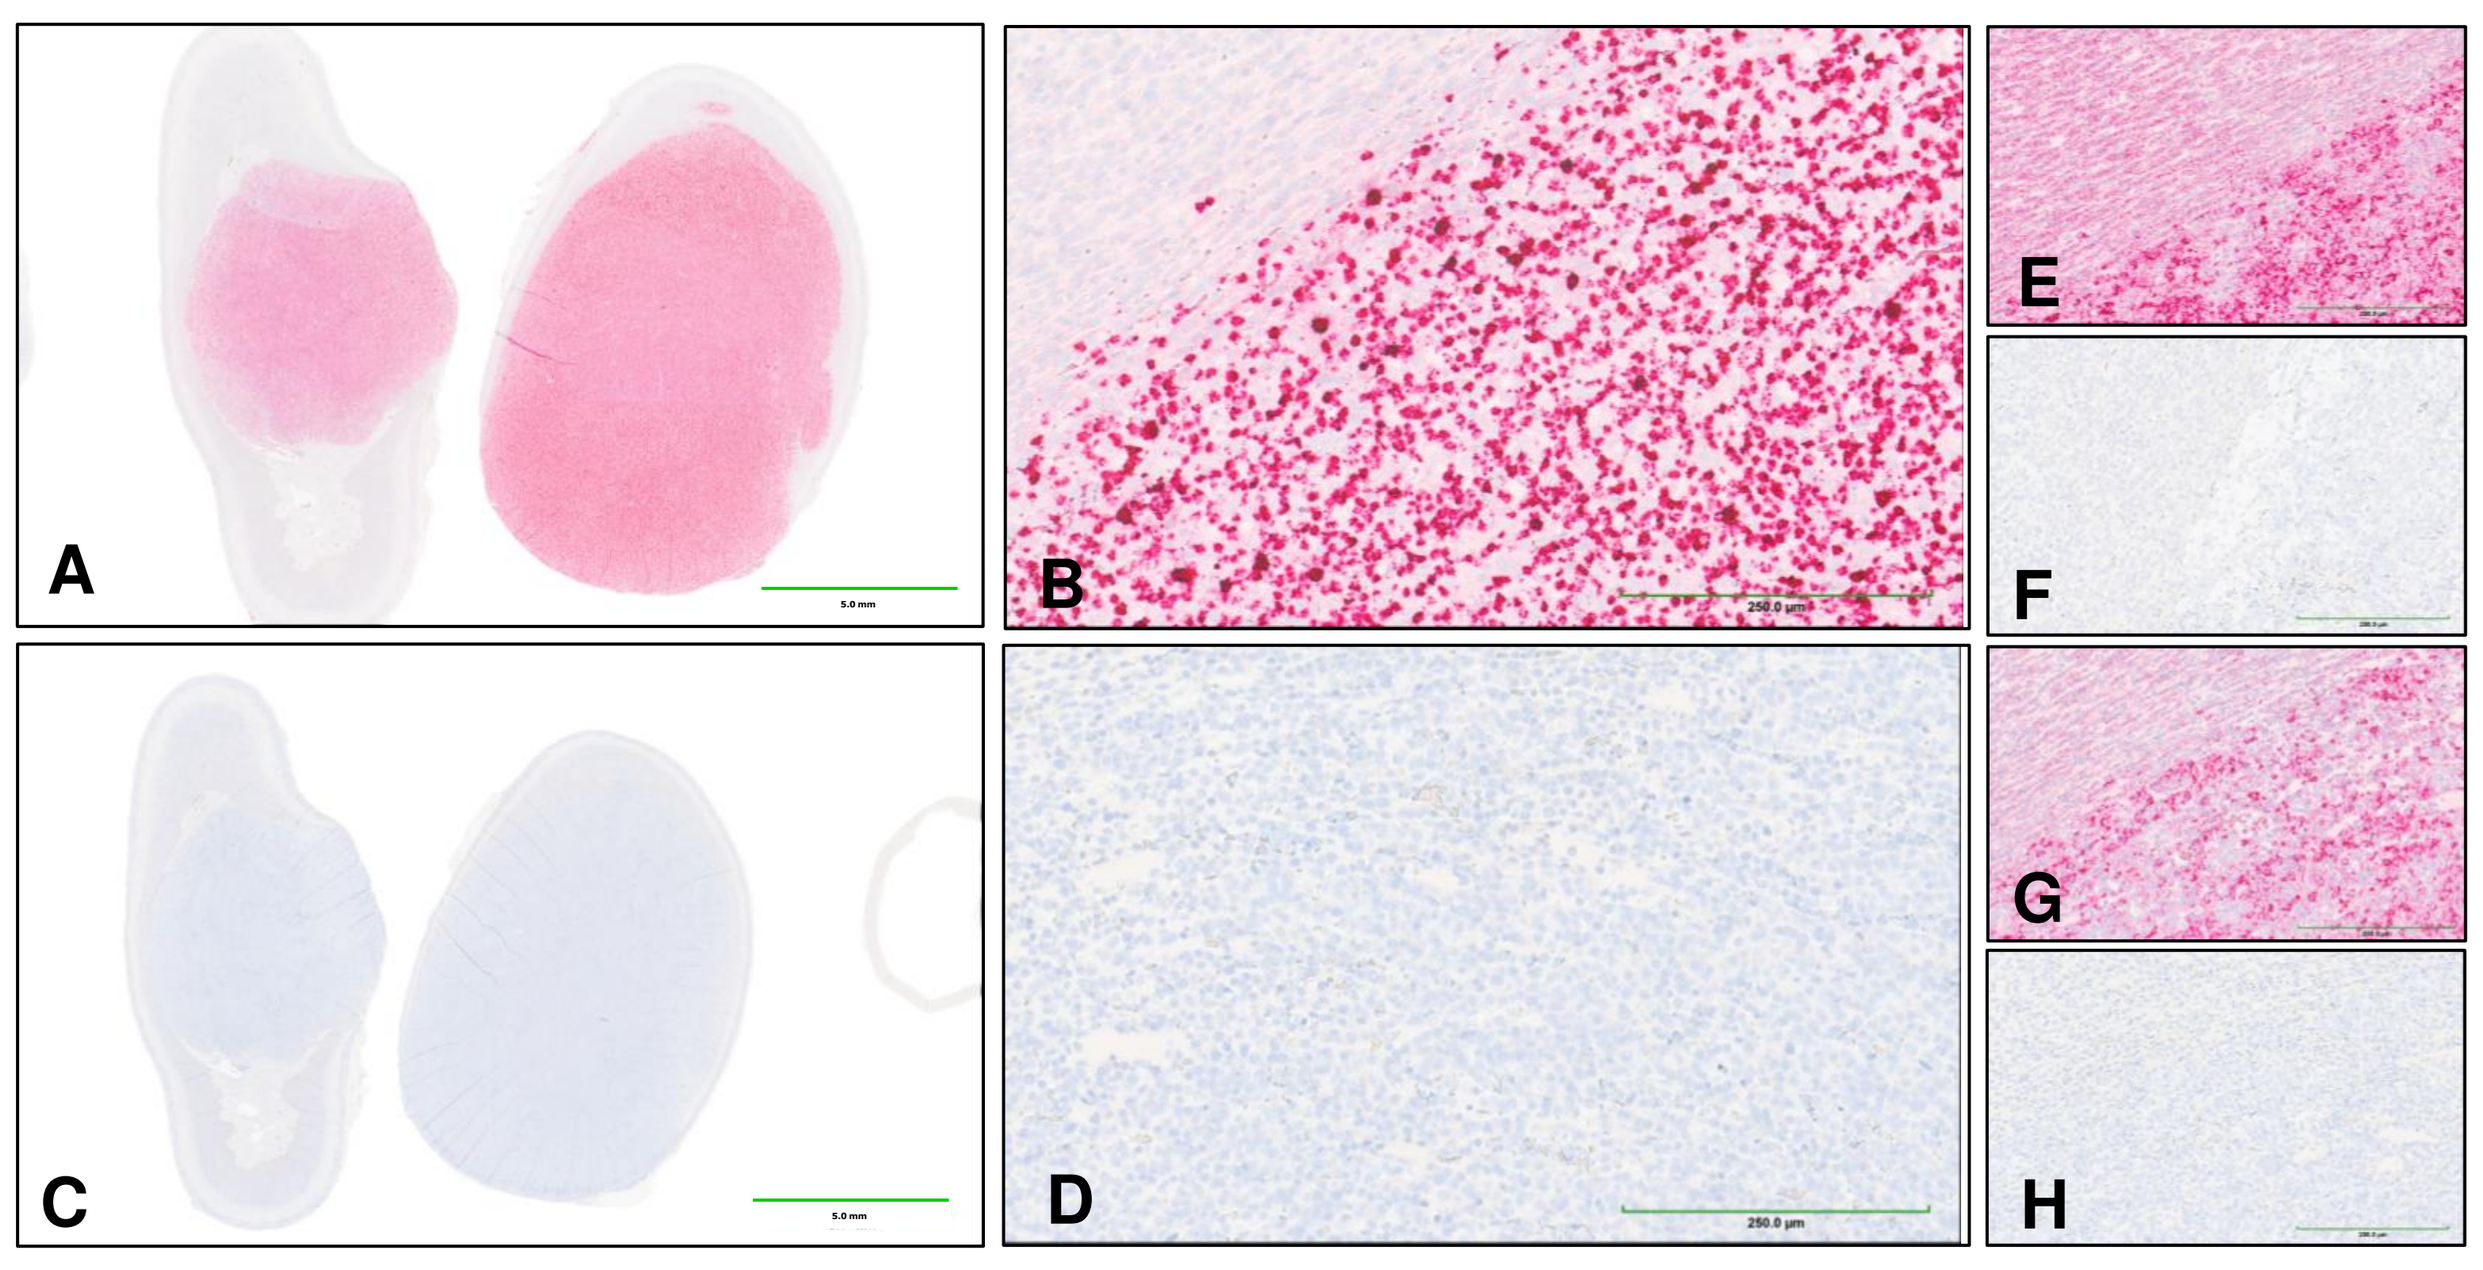
A
B
E
F
C
D
G
H

Figure 4. Detection of RV and LCV in the tumors by ISH

Photomicrographs of a lymphomatous lesion in the adrenal gland assessed by RNAscope® technology ISH for RV (A, B) and LCV (C, D) genomes. RV genome is readily detected in neoplastic cells (*i.e.*, very strong specific staining), the red dots being indicative of RV mRNA signal (A, B). In contrast, LCV mRNA is not detected in the neoplastic tissue (C, D). (E) and (G) show the respective PPIB positive controls for RNA integrity, and (F) and (H) the respective dapB (target absent in the specimen of interest) negative controls.

7. qPCR for herpesviral loads in blood

- **Strong, time-related increases in RV loads** observed from Week 16 in both high dose animals diagnosed with B-cell lymphoma and sacrificed prematurely
- Increase in RV load (outside range of controls) observed from Week 24 in the high dose animal sacrificed as scheduled and diagnosed with B-cell lymphoma
- No changes in RV loads in control animals and other treated animals
- No test article-related changes in LCV and CMV loads in treated animals (*not shown*)

Figure 5. Macaque RV copy number in blood at different timepoints

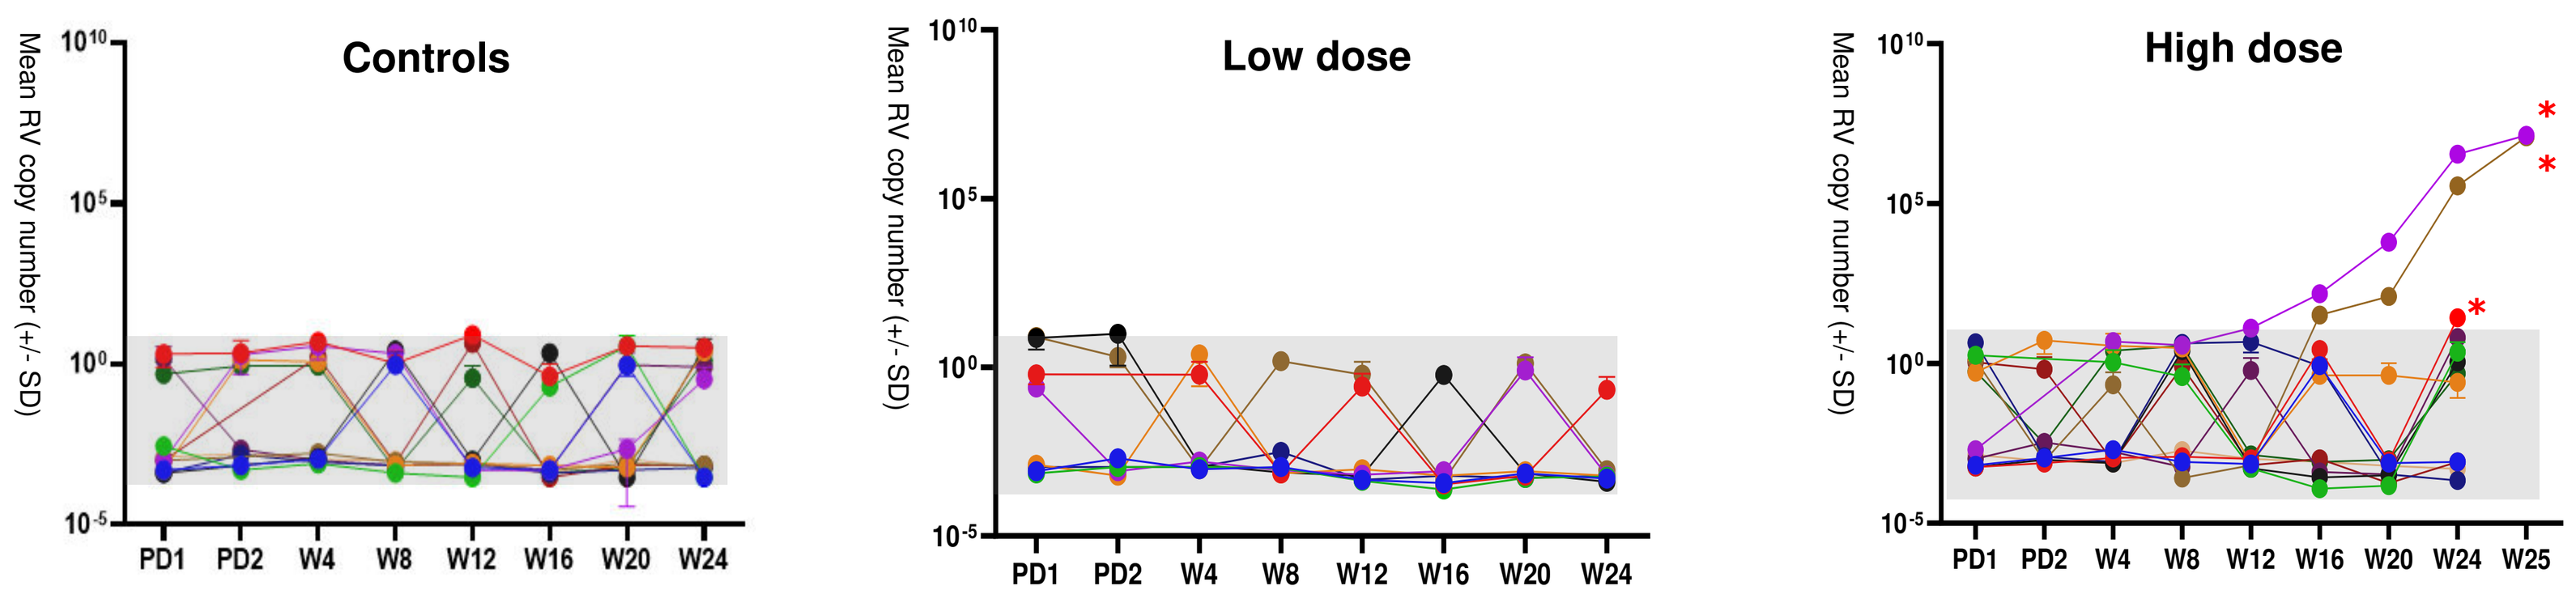
Controls
Low dose
High dose
Mean RV copy number (+/- SD)
10^10
10^5
10^0
10^-5
PD1 PD2 W4 W8 W12 W16 W20 W24
PD1 PD2 W4 W8 W12 W16 W20 W24
PD1 PD2 W4 W8 W12 W16 W20 W24 W25
*

RV loads in the blood from control animals and animals dosed with the test article. The three high dose animals diagnosed with B-cell lymphoma (red asterisks) show increases in macaque RV copy numbers in a time-dependant manner. Copy numbers are estimated using standard curve from RV target DNA sequence in plasmid. PD: Predose. W: Week.

8. Non-neoplastic findings of interest

- At both dose levels, decreased number and size of germinal centers in the spleen, mesenteric lymph node and Peyers' patches
- Opportunistic amoebic infection in the esophagus of the high dose female euthanized prematurely

Figure 6. Opportunistic protozoan infection in a high dose animal

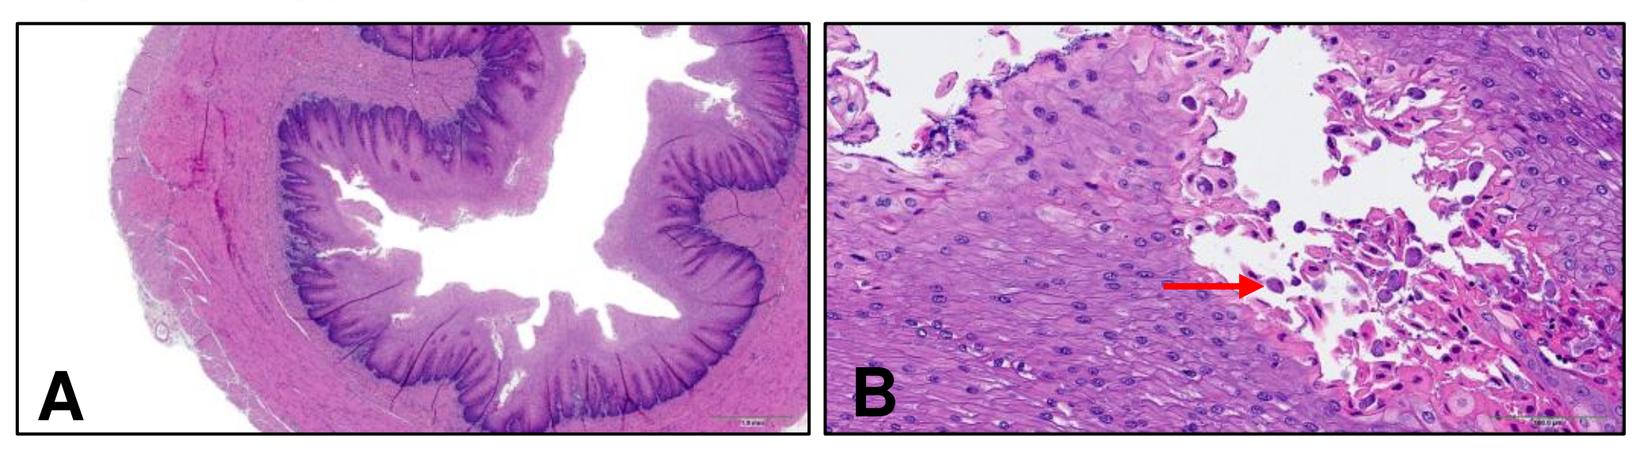
A
B

Photomicrographs of the esophagus at low (A) and high (B) magnifications, showing moderate, diffuse epithelial hyperplasia (A) and intralesional amoebic trophozoites (B; red arrow). H&E staining.

9. Immunophenotyping on blood

- Decreases in central memory and effector memory T helper cells, and effector memory cytotoxic T cells in animals given the test article

10. Serology for RV

- Both males with lymphoma positive at predose, suggesting viral reactivation during treatment
- Female with lymphoma negative at predose and during dosing, suggesting novel infection and no antibody response during treatment
- Caveat: antigen derived from RRV and assay possibly suboptimal for detecting seropositivity against RV in cynomolgus macaques

DISCUSSION & CONCLUSIONS

- The full set of data supports the diagnosis of **macaque RV-associated B-cell lymphomas**, secondary to a profound immunosuppression induced by the chronic administration of the test article.
- The tumors were not due to reactivation of a latent LCV infection.
- RRV is known to establish a latent infection in B lymphocytes of rhesus macaques, and can cause B cell lymphoproliferative diseases in SIV-infected animals.
- One of the major factors involved in RRV-related disease is viral interleukin 6, a cytokine homologue that directly promotes B cell proliferation.
- While lymphoproliferative disease in old world NHPs has most often been reported to be associated with the primate LCV in an immunocompromised context, this report demonstrates that macaque RV can be involved in B-cell lymphomagenesis in animals chronically dosed with an immunomodulatory drug.
- **Opportunistic rhadinoviral infection** should be considered as a differential etiological diagnosis of B-cell lymphomas encountered in NHP safety studies.

SELECT REFERENCES

1. Bergquam EP, Avery N, Shiigi SM, et al. Rhesus rhadinovirus establishes a latent infection in B lymphocytes in vivo. J Virol. 1999 Sep;73(9):7874-6.
2. Bruce AG, Bakke AM, Thouless ME, Rose TM. Development of a real-time QPCR assay for the detection of RV2 lineage-specific rhadinoviruses in macaques and baboons. Virol J. 2005 Jan 5;2:2.
3. Bruce AG, Bielefeldt-Ohmann H, Barcy S, et al. Macaque homologs of EBV and KSHV show uniquely different associations with simian AIDS-related lymphomas. PLoS Pathog. 2012;8(10):e1002962.
4. Estep RD, Wong SW. Rhesus macaque rhadinovirus-associated disease. Curr Opin Virol. 2013 Jun;3(3):245-50.
5. Orzechowska BU, Powers MF, Sprague J, et al. Rhesus macaque rhadinovirus-associated non-Hodgkin lymphoma: animal model for KSHV-associated malignancies. Blood. 2008 Nov 15;112(10):4227-34.
6. Wachtman L, Mansfield K. Viral Diseases of Nonhuman Primates. In Nonhuman Primates in Biomedical Research, Volume 2: Diseases (Abee CR, Mansfield K, Tardif S, Morris T, eds), 2nd ed., 2012; 23-24. Academic Press, London, UK.

ANIMAL CARE AND USE

Performed in accordance with applicable animal welfare regulations at an AAALAC international accredited animal program

DISCLOSURES

Sébastien Laurent, Ambroise Garry, Cris Kamperschroer, Carolin Schramm and Fatima Tariq are employees of Sanofi and may hold shares and/or stock options in the company. Frédéric Gervais, Svenja R. Hartung, Marc Niehoff, and Annette Romeike declare no competing interests.

FUNDING

Study funded by Sanofi

2025 ESTP Annual Meeting

Contact details: sebastien.laurent2@sanofi.com

labcorp sanofi charles river

Poster 24